Radiographic and clinical progression after motor diagnosis in HD is a function of initial striatal volume and functional scores: a re-analysis of TRACK-HD/TRACK-ON HD MRI images and data

Sarah J. Tabrizi, MD, PhD[1], Kirsi M. Kinnunen, PhD[2], Doug Langbehn, MD, PhD[3], Ricardo E. Dolmetsch, PhD[4] , Talaha M. Ali, MD[4] , David L. Cooper, MD, MBA[4]
[1]Huntington's Disease Centre, UCL Institute of Neurology, University College London, London UK; [2] IXICO, London UK; [3] Department of Psychiatry, University of Iowa, Iowa City, IA, USA; Affiliation: [4]uniQure,Lexington, MA, USA

INTRODUCTION

Clinical intervention in Huntington's Disease (HD) is most likely to be effective early in the course of the disease before significant brain atrophy. However, clinical trials in these early patients are complicated by the lack of data on the rate of disease progression in both radiological and clinical endpoints. In earlier populations, biomarkers become more important; volumetric MRI (vMRI) methods continue to evolve. Newer approaches to disease modification rely on a minimum striatal volume to identify a population with greater benefit and lower surgical risk; little is known about impact of initial volume on clinical and radiographic progression Here we describe a collaboration between CHDI, uniQure and IXICO on a pilot vMRI study looking at reanalysis of participants with early clinically-diagnosed HD from TRACK-HD/TRACK-ON using contemporary vMRI algorithms.

OBJECTIVE

To reanalyze radiographic and clinical progression in participants with early clinically-diagnosed HD from TRACK-HD/TRACK-ON HD studies[1,2].

METHODS

From the TRACK-HD/TRACK-ON HD studies, participants were identified with Diagnostic Confidence Level (DCL) =4 and Total Functional Capacity (TFC) 9-13 at baseline or subsequent visits, with at least 1 evaluable follow-up MRI scan. All baseline vMRI parameters were assessed using LEAP (Learning Embeddings for Atlas Propagation[3]) with longitudinal assessment of whole brain, ventricles and caudate using LLEAP[4] and cross-sectional assessment of putamen using a novel CNN (convolutional neural network) method[5].

RESULTS

From 156 participants meeting the clinical criteria (476 MRI scans), 120 had 1-5 years of follow-up (412 analyzable scans); 15 participants had baseline scans that failed image QC.

- Of 105 remaining participants, 74 had low striatal volume (< 2.5 cm^3 putamen, < 2.0 cm^3 caudate – per side) and 31 had high (≥ 2.5 cm^3 putamen, ≥ 2.0 cm^3 caudate – per side).
- Changes in vMRI and clinical progression were stratified by subgroups of TFC: 12-13 vs. 9-11; striatal volume: high vs low; and TMS: ≤ 25 vs >25.
- The distribution of baseline TMS in participants with high striatal volumes are shown in **Figure 1**.
- 41% of those with TMS ≤ 25 had initial striatal volume in the high range, and only 6% of those with TMS > 25 at baseline had baseline striatal volume in the high range.

Figure 1. Baseline TMS in participants with high striatal volumes

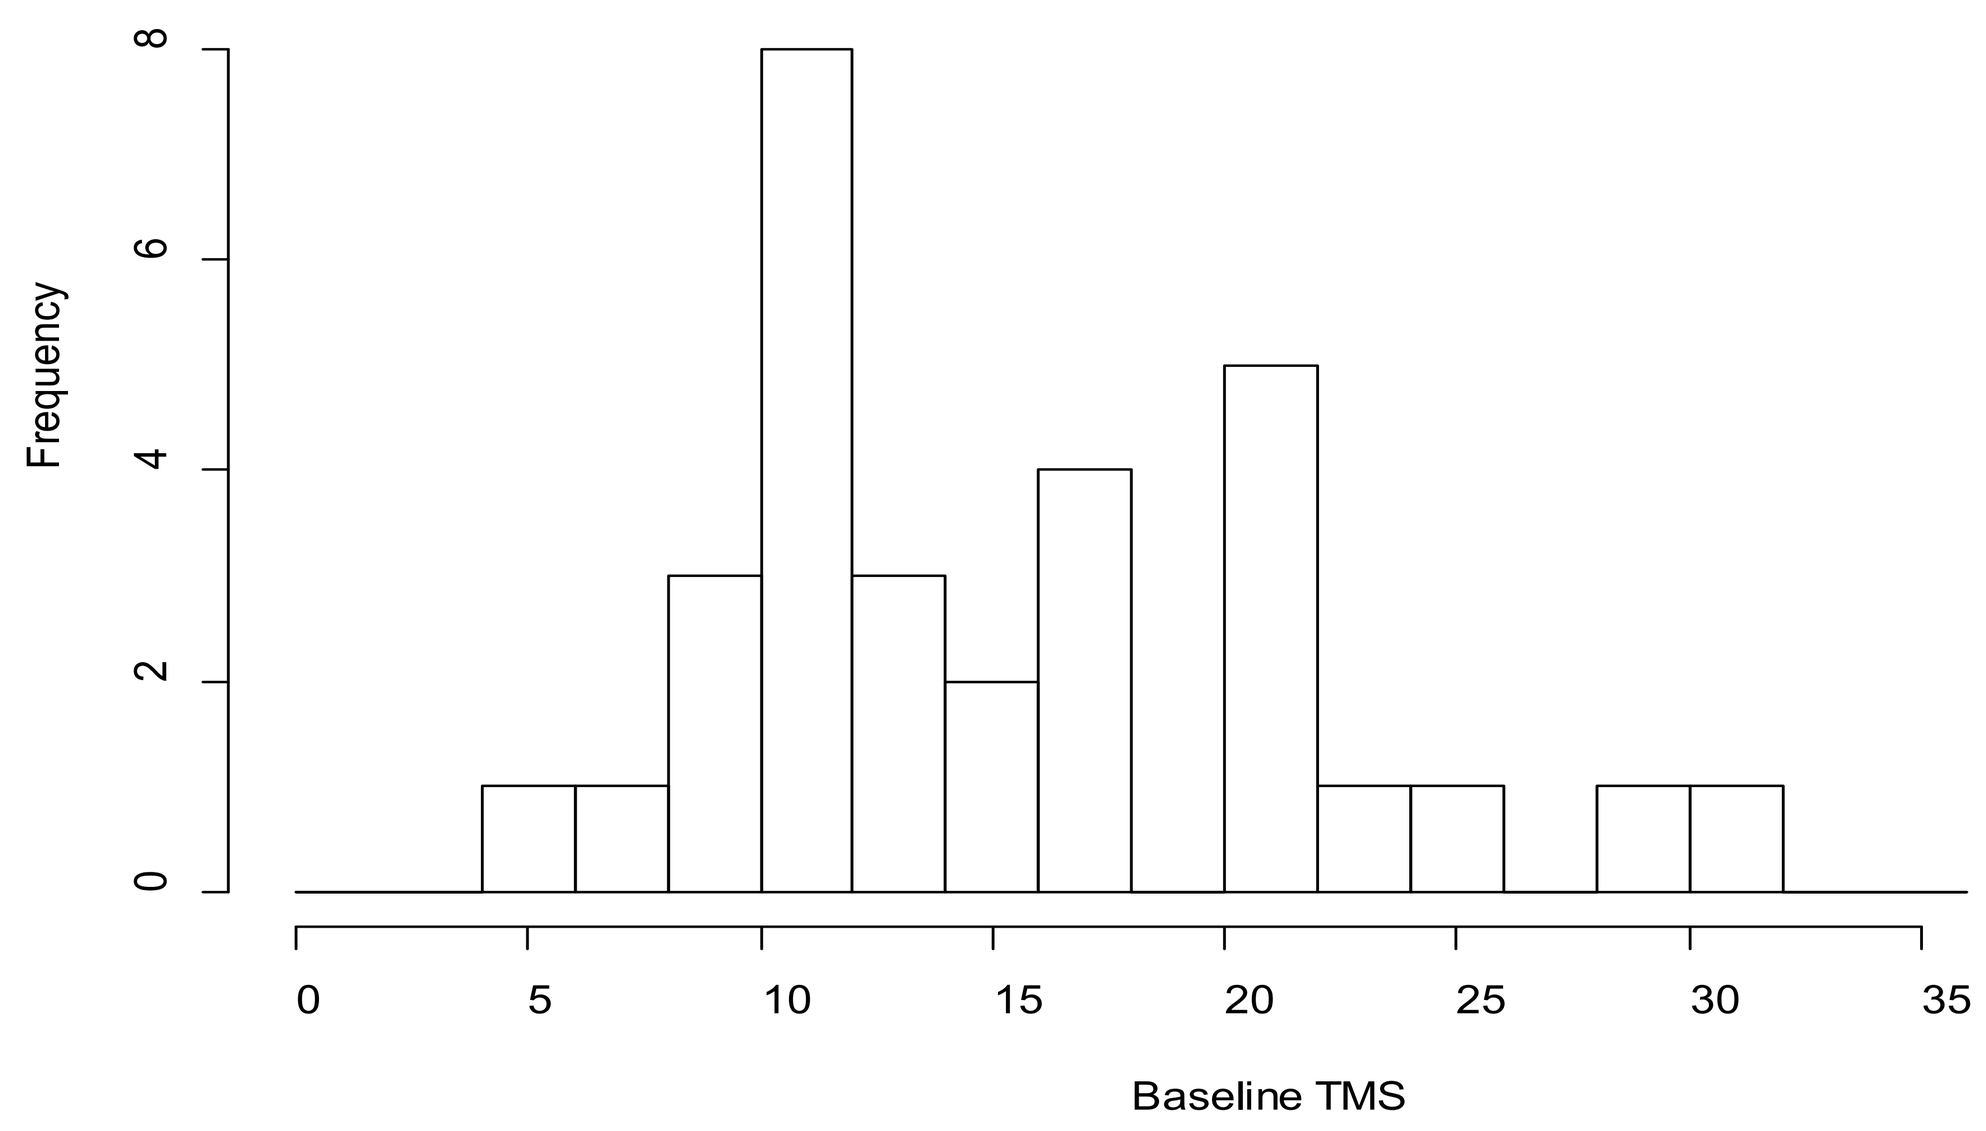
Frequency
Baseline TMS

Table 1. TRACK-HD vMRI changes by baseline TFC, vMRI striatal volume and TMS (NS; p>0.050)

	Rate of change % per year	Initial TFC 12-13 vs 9-11	Initial vMRI High vs Low SV	Initial TMS ≤ 25 vs >25
Whole Brain (atrophy)	0.92% p<0.0001	0.95% vs 0.88% p=0.554 (NS)	0.75% vs 0.97% p=0.058 (NS)	0.90% vs 0.96% p=0.602 (NS)
Ventricles (enlargement)	7.85% p<0.0001	8.71% vs 6.52% p=0.013	7.03% vs 8.40% p=0.120 (NS)	8.00% vs 7.50% p=0.120 (NS)
Caudate (atrophy)	4.96% p<0.0001	5.00% vs 4.90% p=0.798 (NS)	4.00% vs 5.60% p<0.0001	4.70% vs 5.60% p=0.039
Putamen (atrophy)	3.22% p<0.0001	3.24% vs 3.21% p=0.925 (NS)	3.00% vs 3.40% p=0.222 (NS)	3.20% vs 3.30% p=0.725 (NS)
Hippocampus (atrophy)	0.88% p<0.0001	0.93% vs 0.83% p=0.447	0.70% vs 0.96% p=0.081 (NS)	0.77% vs 1.12% p=0.013

Table 2. TRACK-HD clinical progression by baseline TFC, vMRI striatal volume and TMS (NS; p>0.050)

	Rate of change points per year	Initial TFC 12-13 vs 9-11	Initial vMRI High vs Low SV	Initial TMS ≤ 25 vs >25
TFC (decrease)	0.51 p<0.0001	0.58 vs 0.42 p=0.278 (NS)	0.46 vs 0.60 p=0.42 (NS)	0.41 vs 0.75 p=0.035
TMS (increase)	2.38 p<0.0001	2.27 vs 2.48 p=0.738 (NS)	1.40 vs 2.87 p=0.044	2.32 vs 2.43 p=0.870 (NS)
SDMT (decrease)	1.70 p<0.0001	2.00 vs 1.20 p=0.046	1.21 vs 1.82 p=0.177 (NS)	1.41 vs 2.34 p=0.032
SWR (decrease)	2.79 p<0.0001	2.63 vs 3.04 p=0.602 (NS)	1.74 vs 3.19 p=0.095 (NS)	2.44 vs 3.77 p=0.114 (NS)
cUHDRS	0.74 p<0.0001	0.76 vs 0.72 p=0.818 (NS)	0.51 vs 0.87 p=0.018	0.63 vs 1.00 p=0.007

SDMT; Symbol Digit Modality Test, SWR; Stroop Word Reading, cUDHRS; composite Unified Huntington's Disease Rating Scale

- Annualized percentage change in vMRI was significant (p<0.0001) for whole brain atrophy (0.92%), ventricle enlargement (7.85%), caudate atrophy (4.96%), putamen atrophy (3.22%) and hippocampus atrophy (0.88%) **(Table 1)**.
- Annualized clinical progression was significant (p<0.0001) for TFC (0.51), TMS (2.38), SDMT (1.70), SWR (2.79), and cUHDRS (0.74) **(Table 2)**.
- Both vMRI progression **(Table 1)** and clinical progression **(Table 2)** were slower across all parameters for subjects with high initial striatal volume and lower TMS.

Figure 2. TRACK-HD MRI progression by baseline striatal volume (SV)

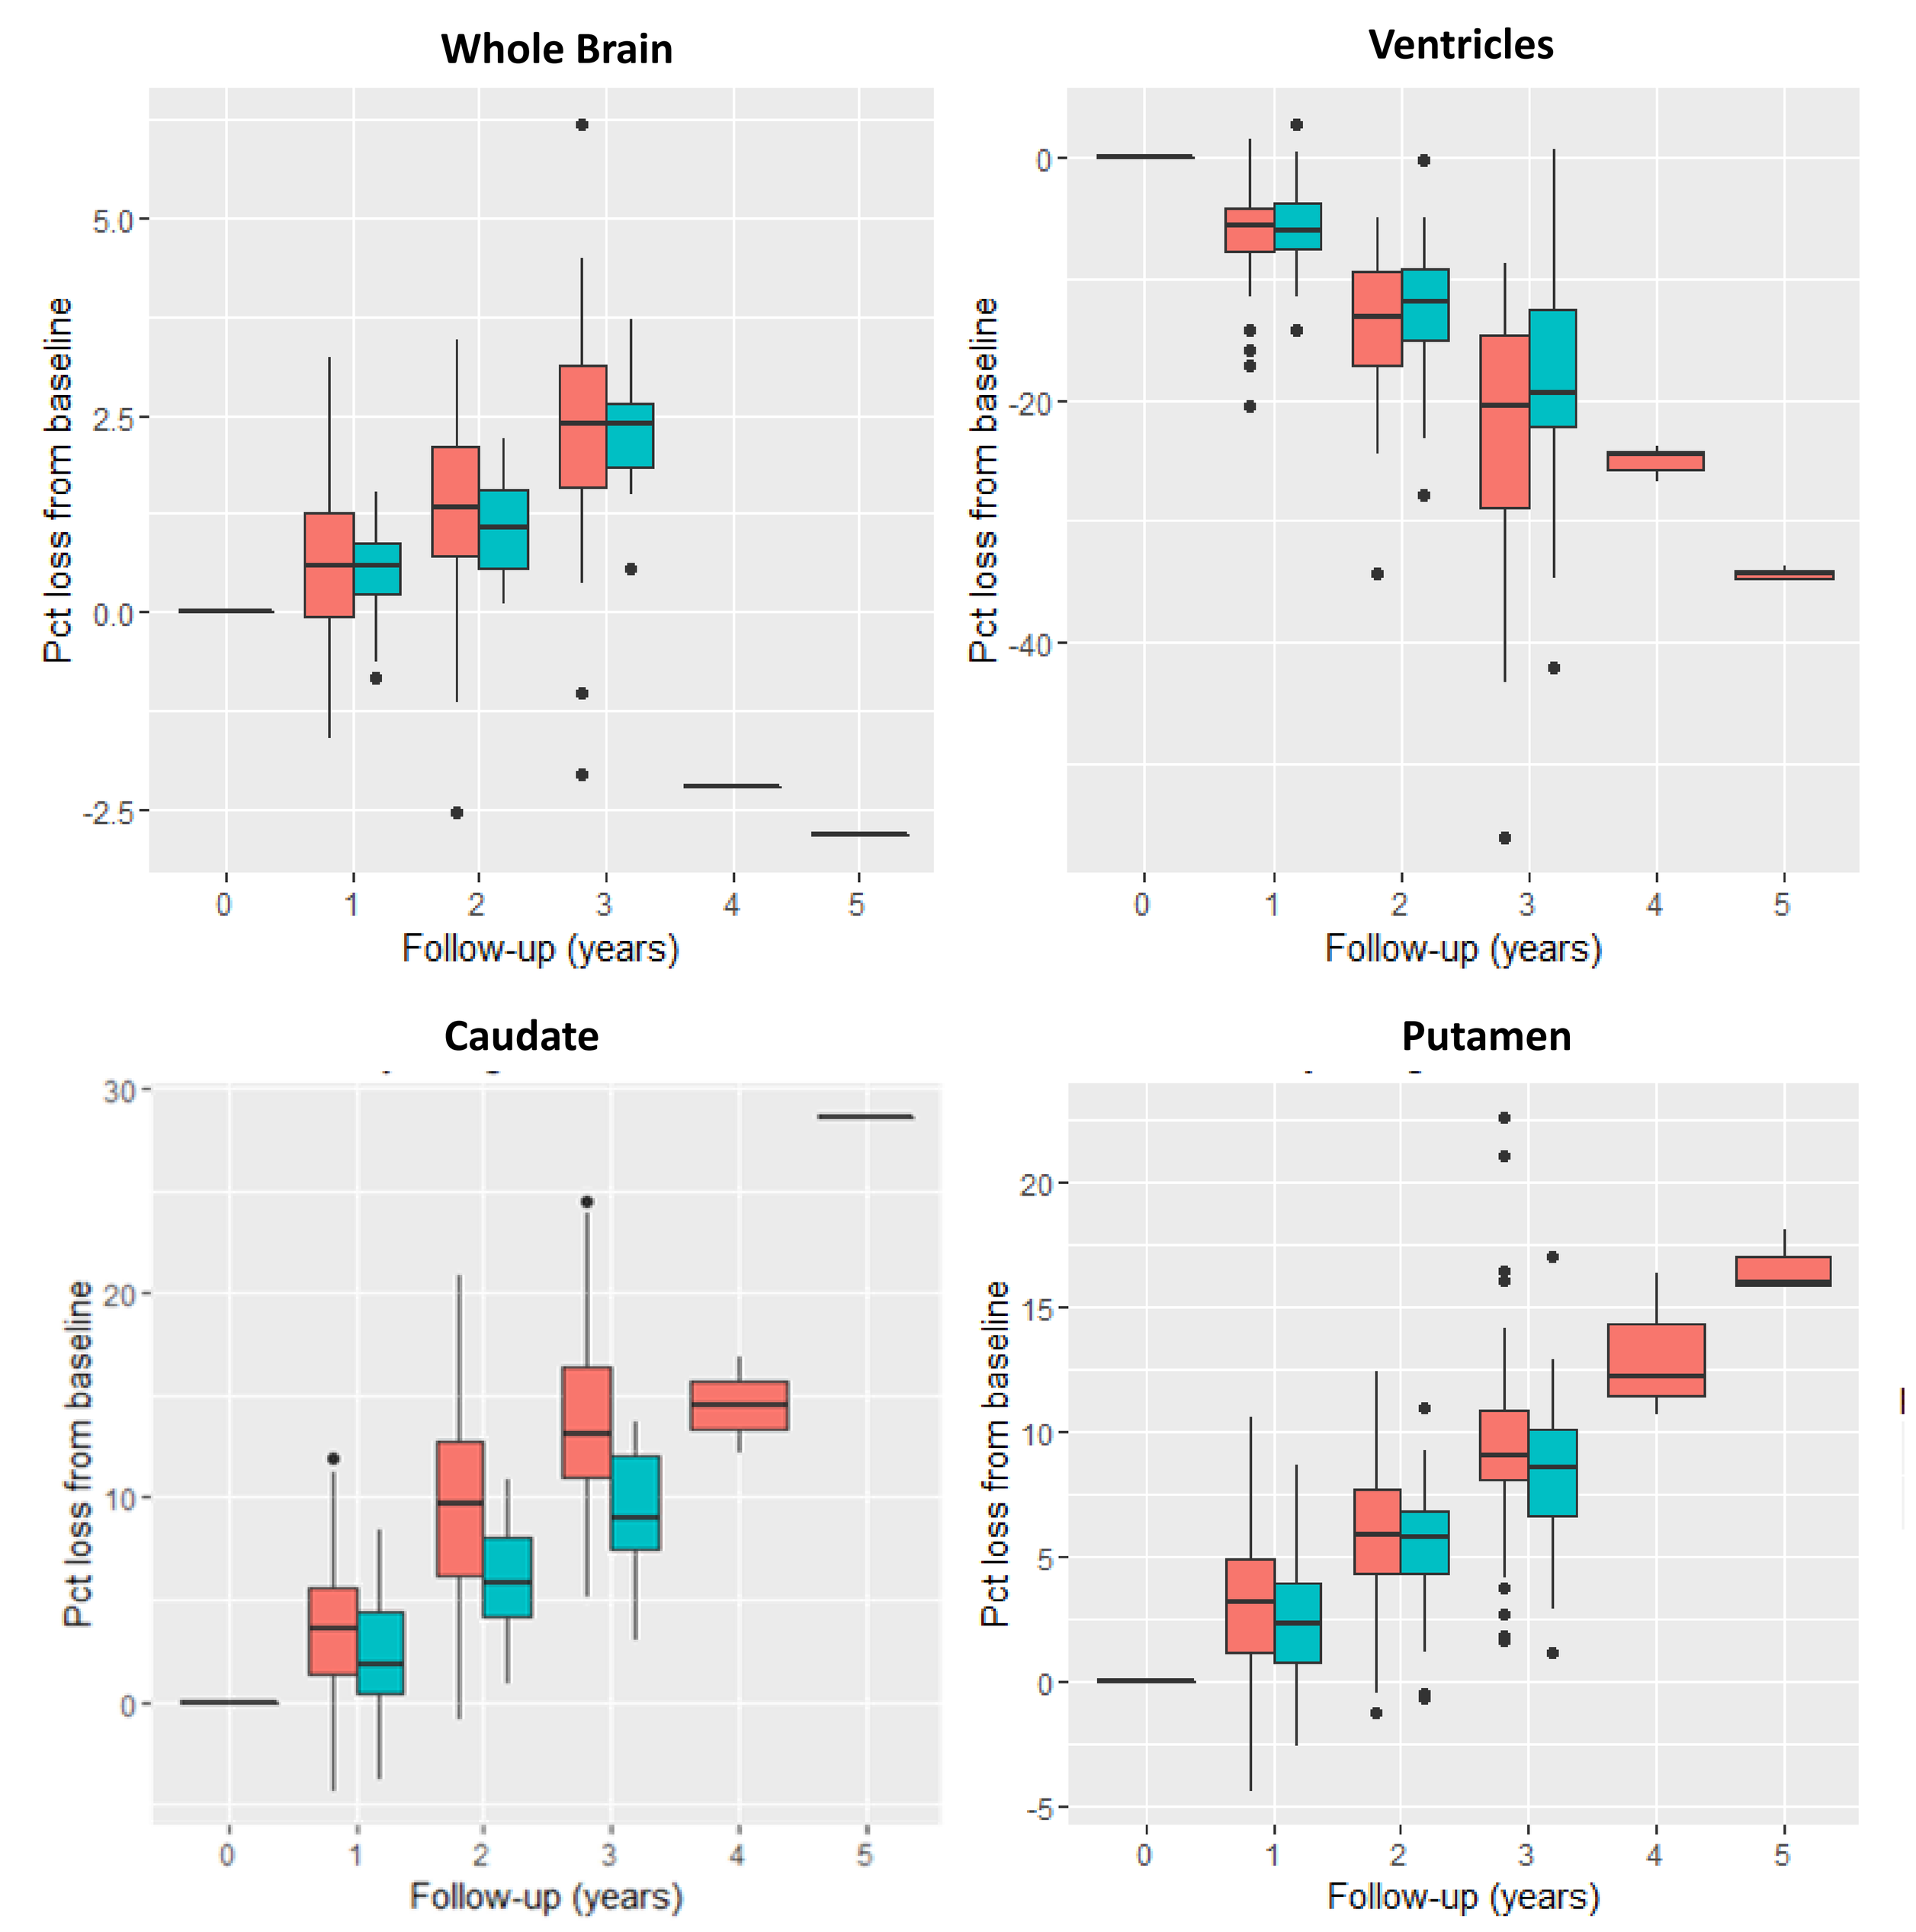
Low SV
High SV
Whole Brain
Ventricles
Caudate
Putamen
Pct loss from baseline
Follow-up (years)

Figure 3. TRACK-HD clinical progression by baseline striatal volume (SV)

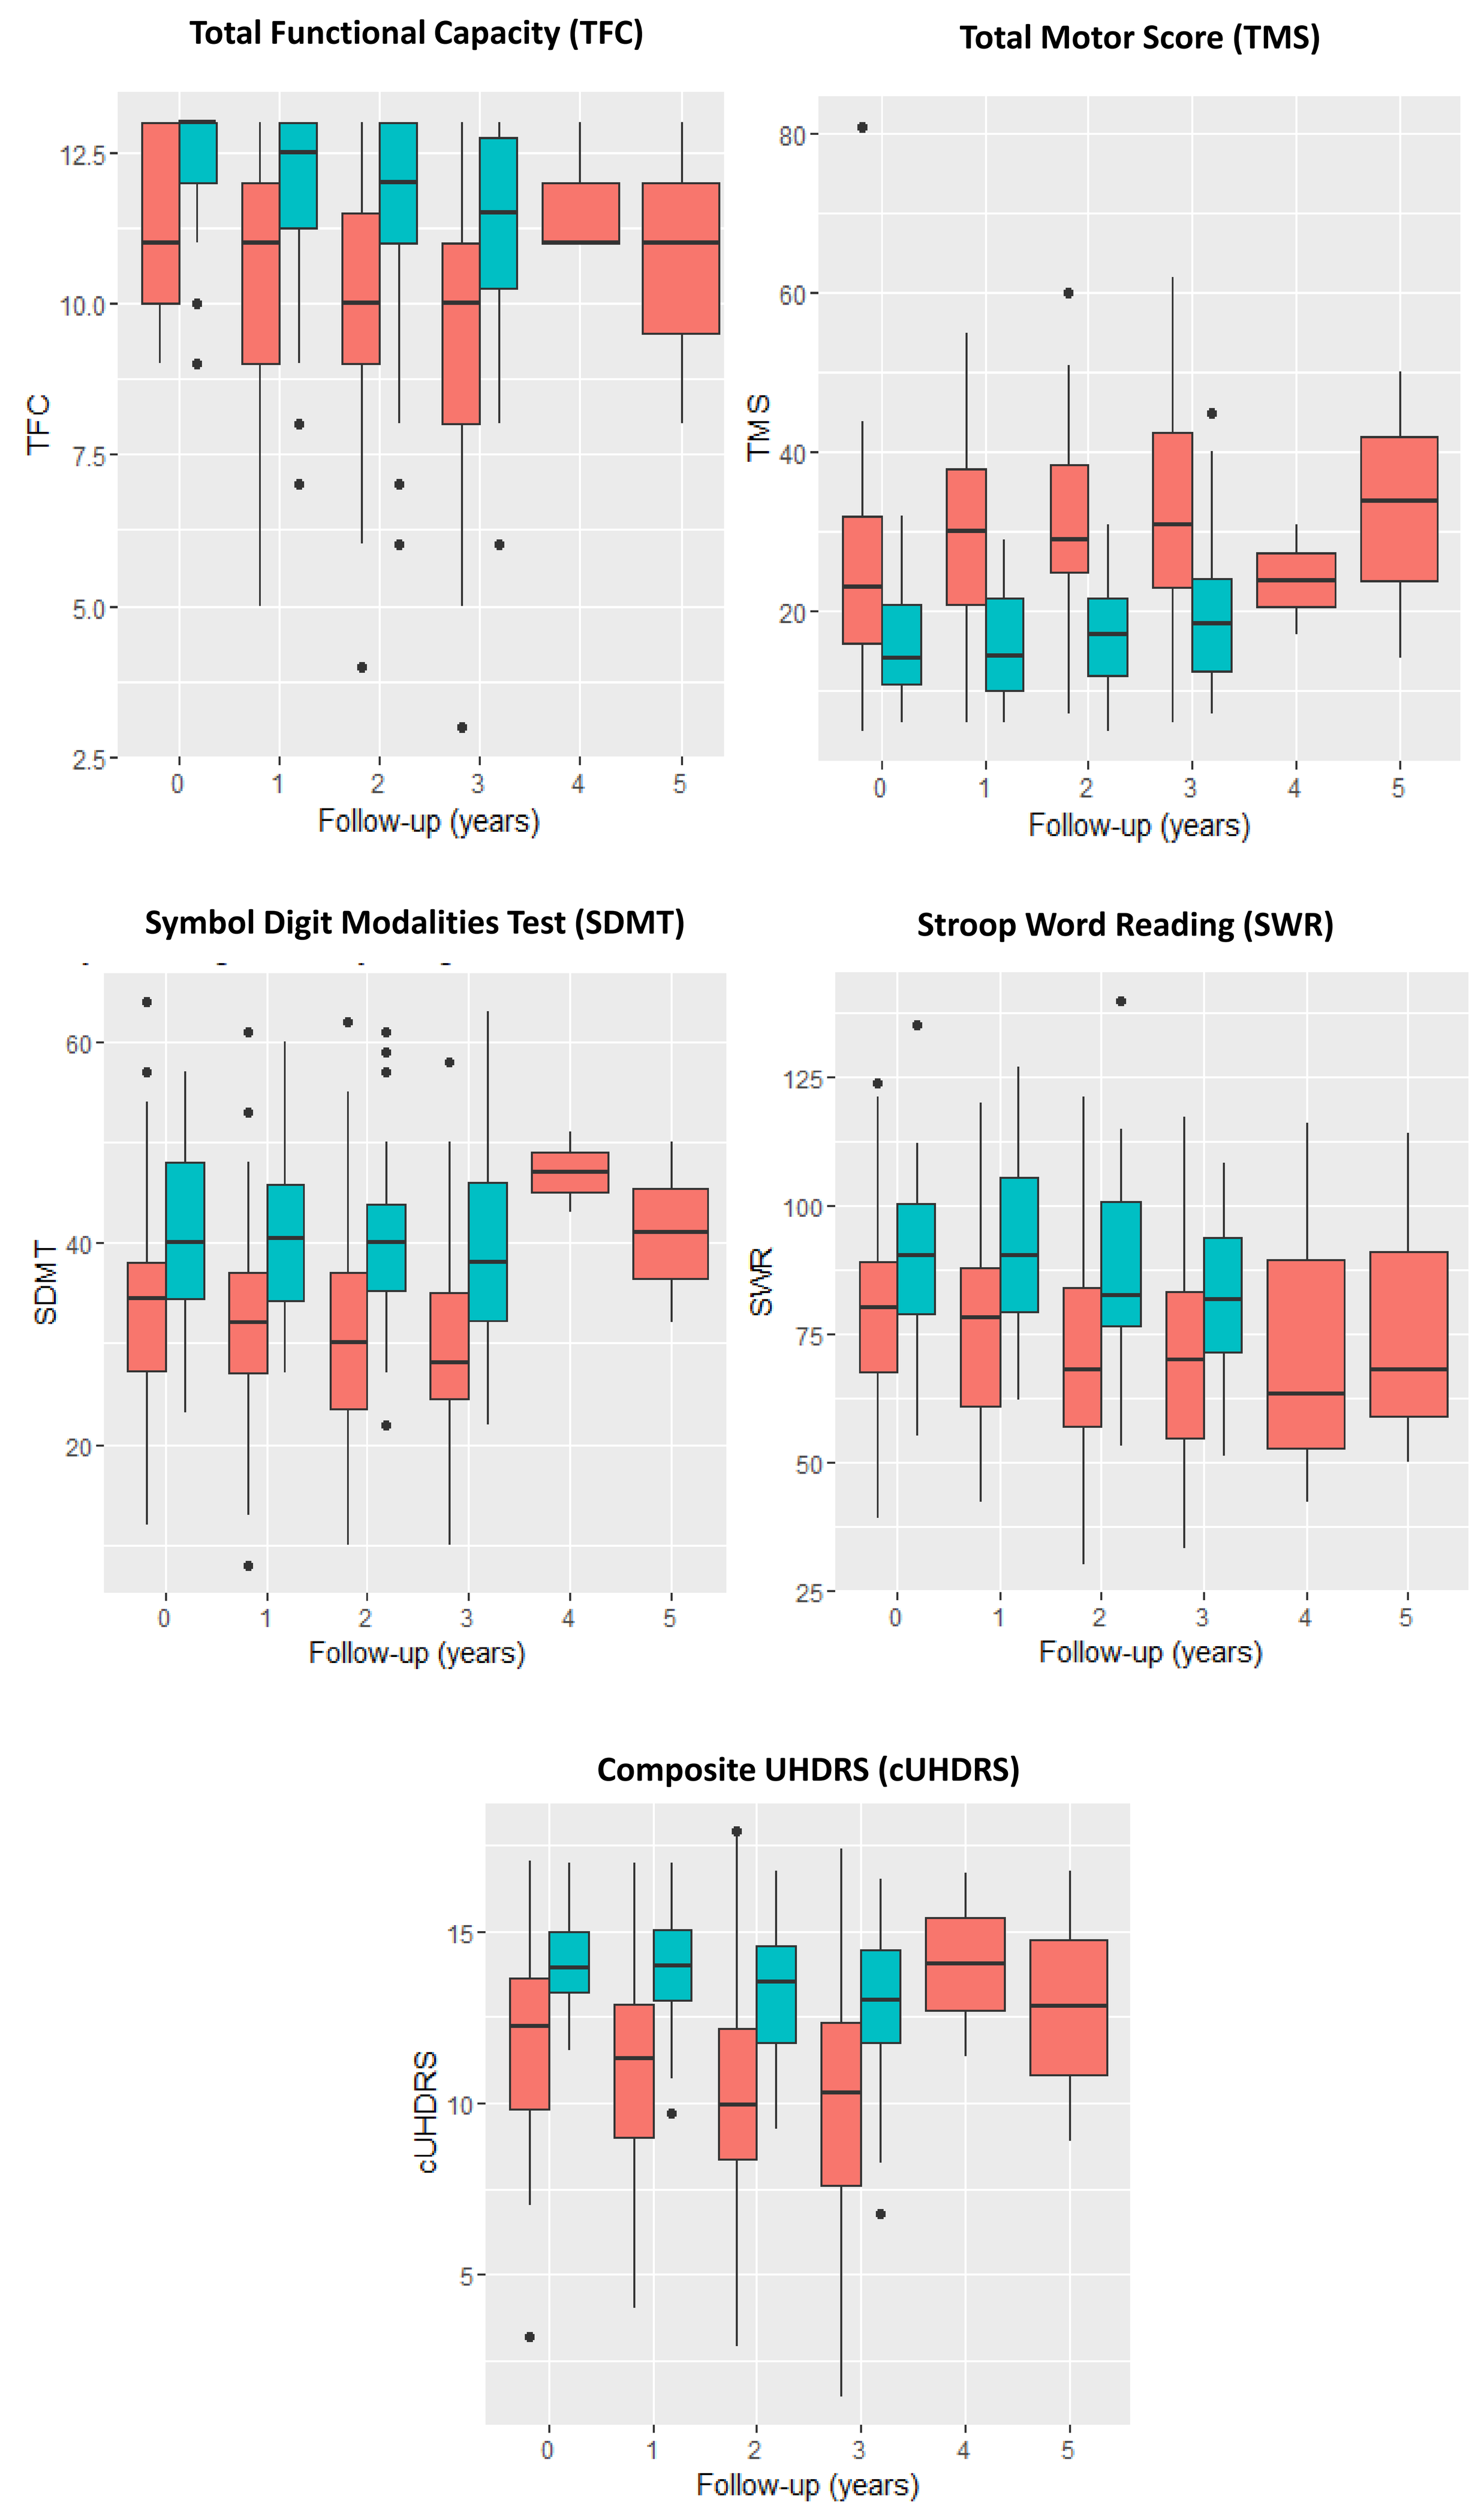
Low SV
High SV
Total Functional Capacity (TFC)
Total Motor Score (TMS)
Symbol Digit Modalities Test (SDMT)
Stroop Word Reading (SWR)
Composite UHDRS (cUHDRS)
Follow-up (years)

- Based on initial high vs low striatal volume, vMRI percentage change was significant for caudate atrophy at 4.00% vs 5.60% (p<0.0001) **(Table 1, Figure 2)** and change in points was significant for TMS with 1.40 vs 2.87 (p=0.044) and cUHDRS with 0.51 vs 0.87 (p=0.018) **(Table 2, Figure 3).**
- Based on initial TFC 12-13 vs 9-11 vMRI percentage change was significant for ventricle enlargement 8.71% vs 6.52% (p=0.013) and hippocampus atrophy 0.93% vs 0.83% (p=0.447) **(Table 1)** and change in points was significant for SDMT 2.00 vs 1.20 (p=0.046) **(Table 2).**

Figure 4. TRACK-HD MRI progression by baseline total motor score (TMS)

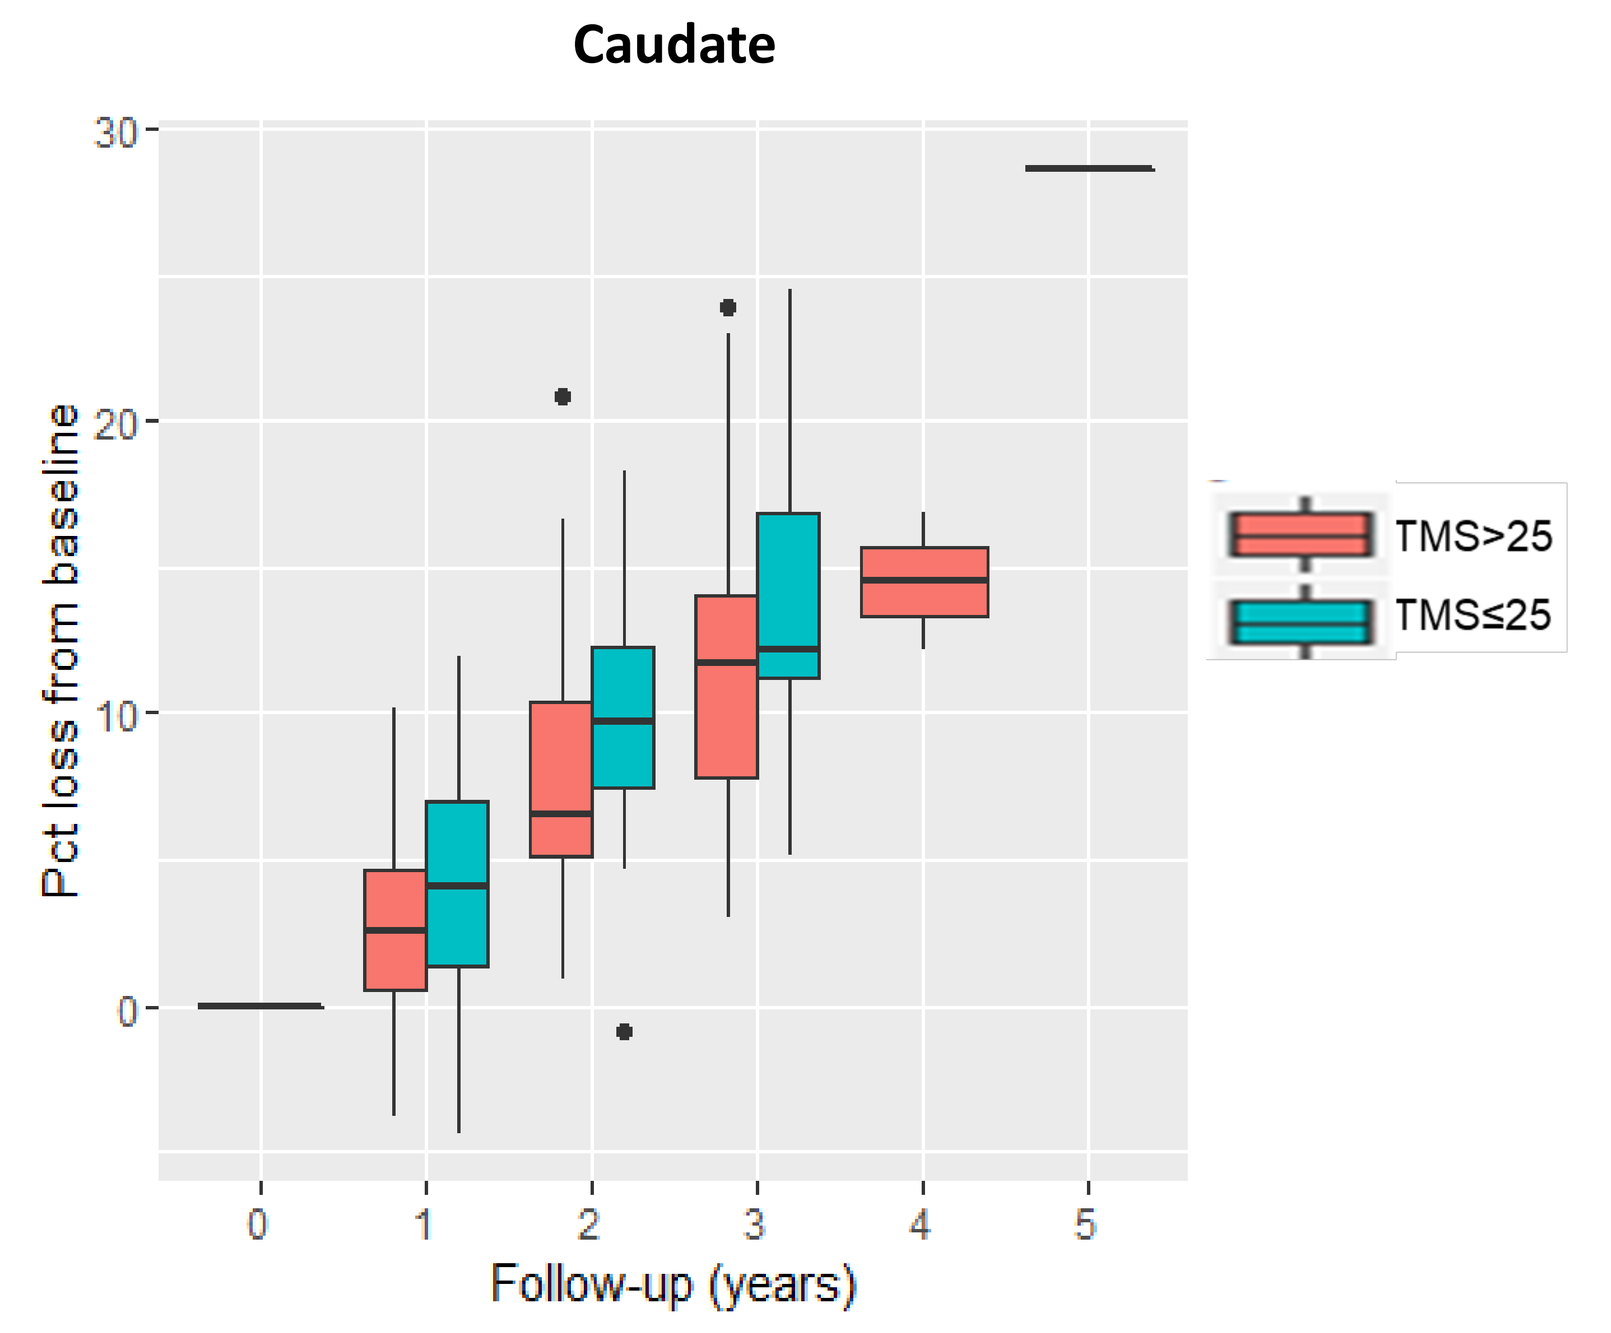
Caudate
Pct loss from baseline
Follow-up (years)
TMS>25
TMS≤25

- Based on initial TMS ≤ 25 vs >25, vMRI percentage change was significant for caudate atrophy 4.70% vs 5.60%; (p=0.039) and hippocampus atrophy 0.77% vs 1.12% (p=0.013) **(Figure 4)** and change in points was significant for TFC was 0.41 vs 0.75 (p=0.035), SDMT 1.41 vs 2.34 (p=0.032), and cUHDRS 0.63 vs 1.00 (p=0.007) **(Table 2).**

Figure 5. TRACK-HD TMS progression of individual participants by baseline TFC

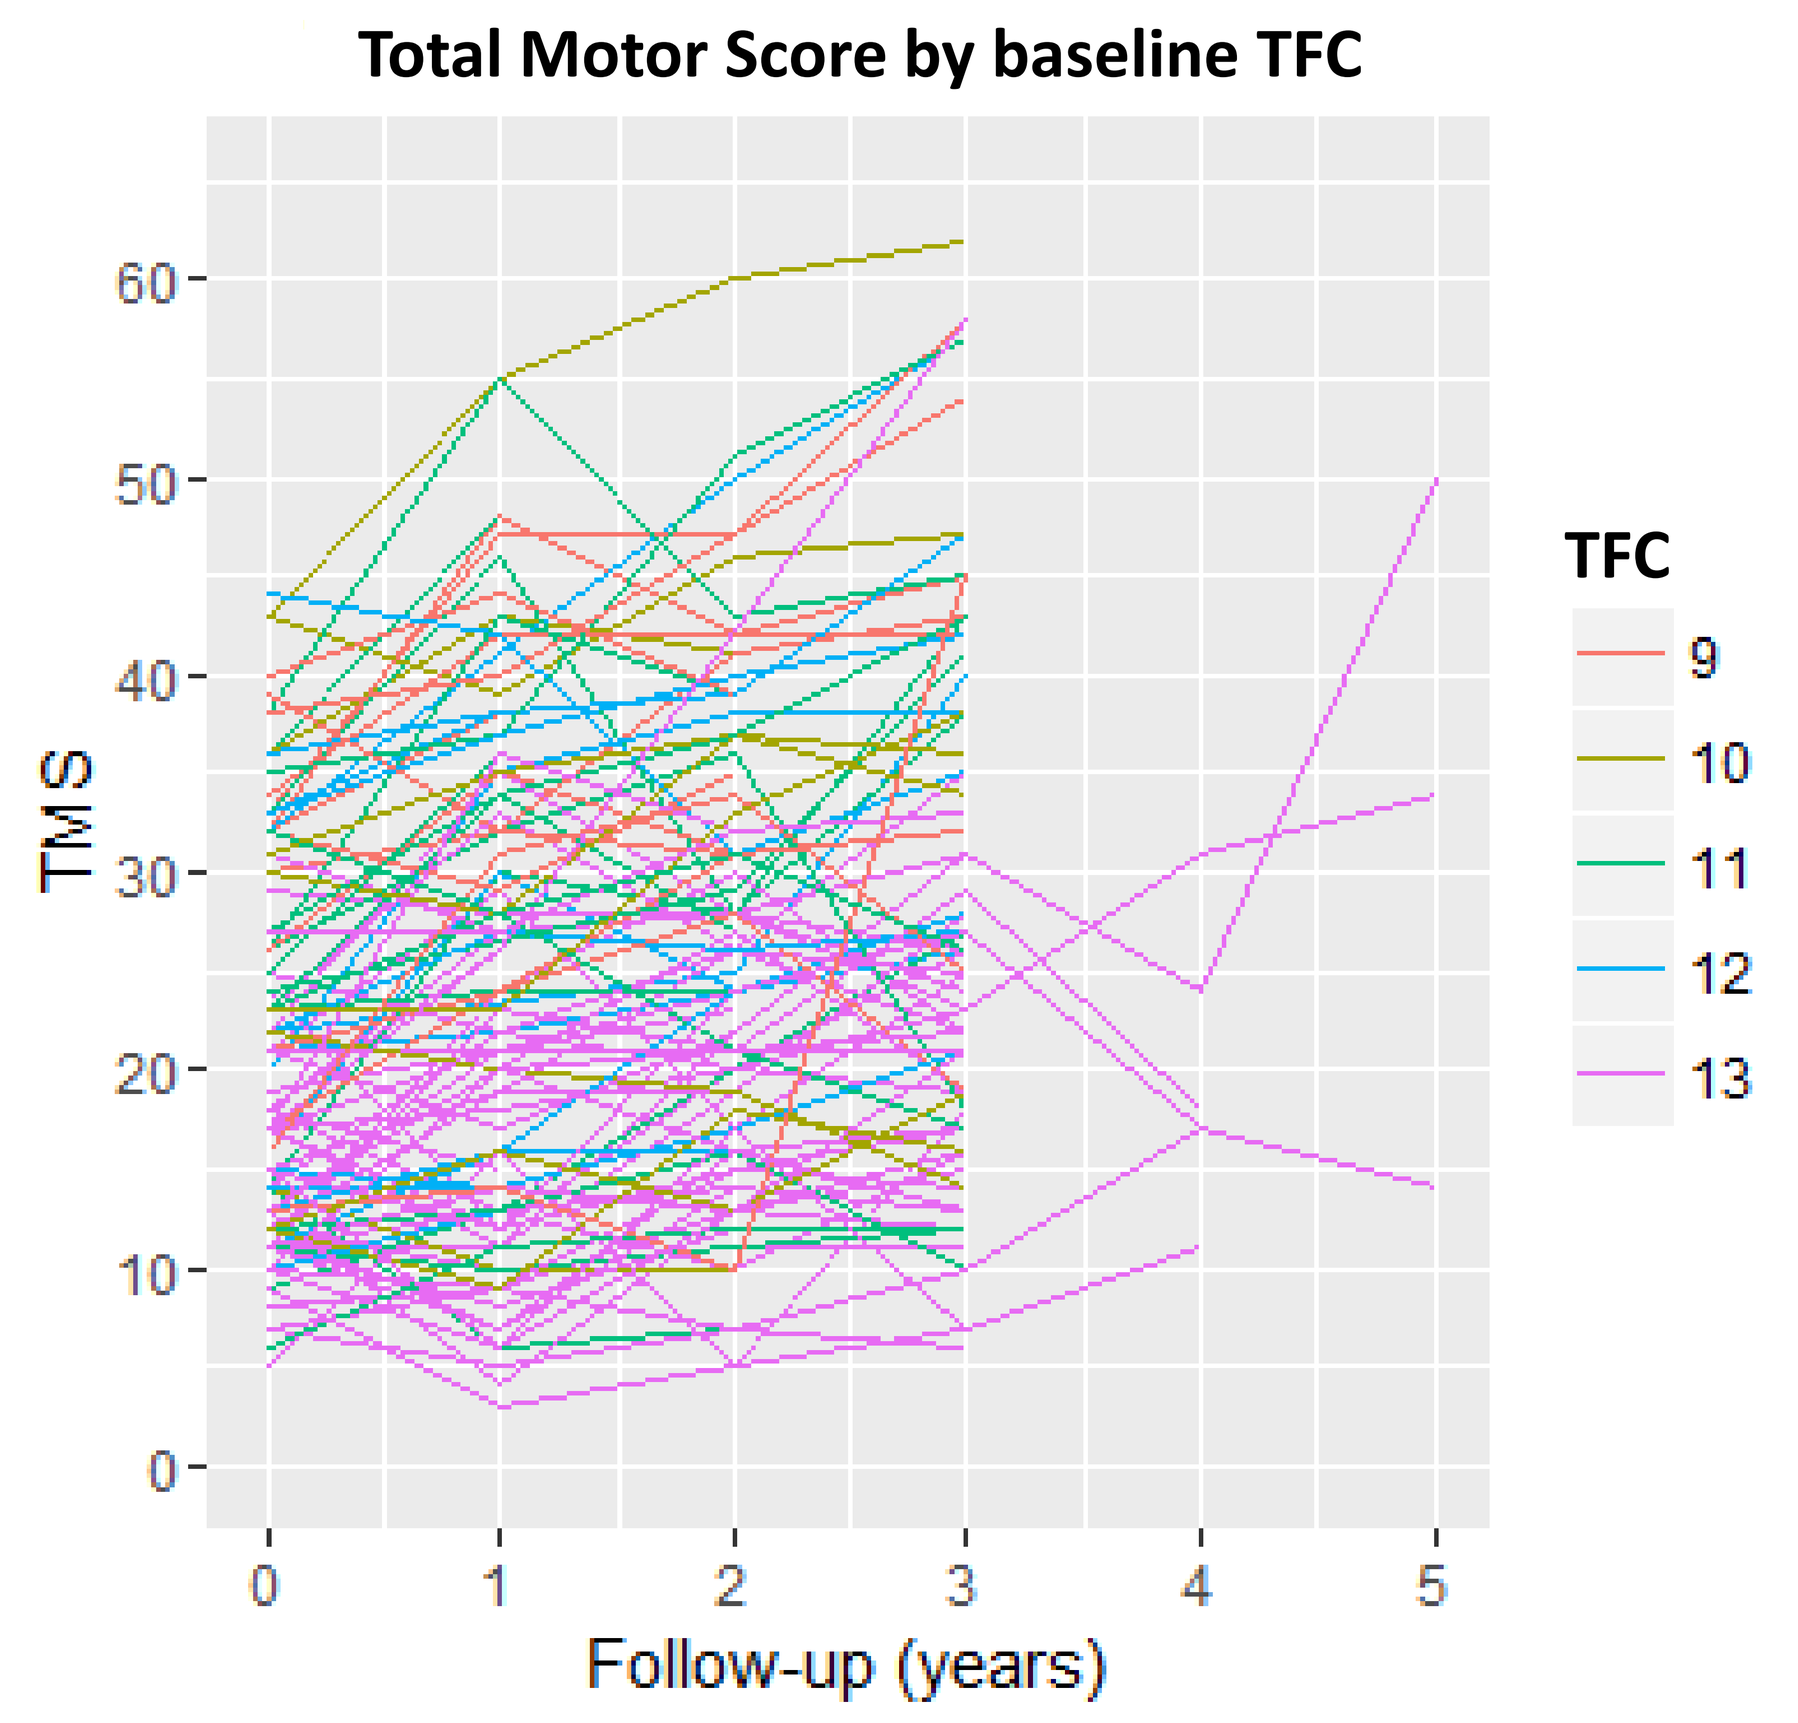
Total Motor Score by baseline TFC
TMS
Follow-up (years)
TFC
9
10
11
12
13

- Regardless of baseline TFC, TMS progression shows substantial year-to-year variability. Although mean scores worsen over several years, apparent improvement between two annual measures is not uncommon **(Figure 5).**

CONCLUSIONS

Using modern vMRI algorithms, this pilot study provides additional insights on annualized radiographic changes and clinical progression in a subset of participants with early clinically-diagnosed HD. For subjects that had high initial striatal volume and lower TMS, clinical and vMRI progression was slower across all parameters.

The finding that after symptoms manifest, the rate of clinical progression (e.g., cUHDRS) and vMRI changes (e.g., caudate atrophy) were impacted more by the extent of initial striatal atrophy and motor symptoms than by functional status, underscores the need to further define factors leading to variability in progression rates in early-stage HD.

REFERENCES

1. Tabrizi SJ, Scahill RI, Owen G, et al; TRACK-HD Investigators. *Lancet Neurol*. 2013;12(7):637-649. doi:10.1016/S1474-4422(13)70088-7
2. Klöppel S, Gregory S, Scheller E, et al; Track-On Investigators. *EBioMedicine*. 2015;2(10):1420-1429. doi:10.1016/j.ebiom.2015.08.002
3. Wolz R, Aljabar P, Hajnal JV et al. *NeuroImage* 2010;49(2):1316-1325. doi:10.1016/j.neuroimage.2009.09.069
4. Wolz R, Heckemann R, Aljabar P et al. *NeuroImage* 2010;52(1):109-118. doi:10.1016/j.neuroimage.2010.04.006
5. Weatheritt et al. *HSG* 2020

Presented at the 17th Annual HD Therapeutics Conference, Palm Springs, CA, February 28–March 3, 2022